A Fluorescent Labeling and Enrichment System for Glycopeptides Generated from Proteolytic Digestion of IgG mAbs; A System That Can Be Used as Part of the Peptide Mapping Workflow.

Jenifer L. Hendel*, Concepción Badía-Tortosa, Daniel I. R. Spencer, Daryl L. Fernandes | *Ludger Ltd, Culham Science Centre, Abingdon, Oxfordshire OX14 3EB, UK*

Email: jenifer.hendel@ludger.com

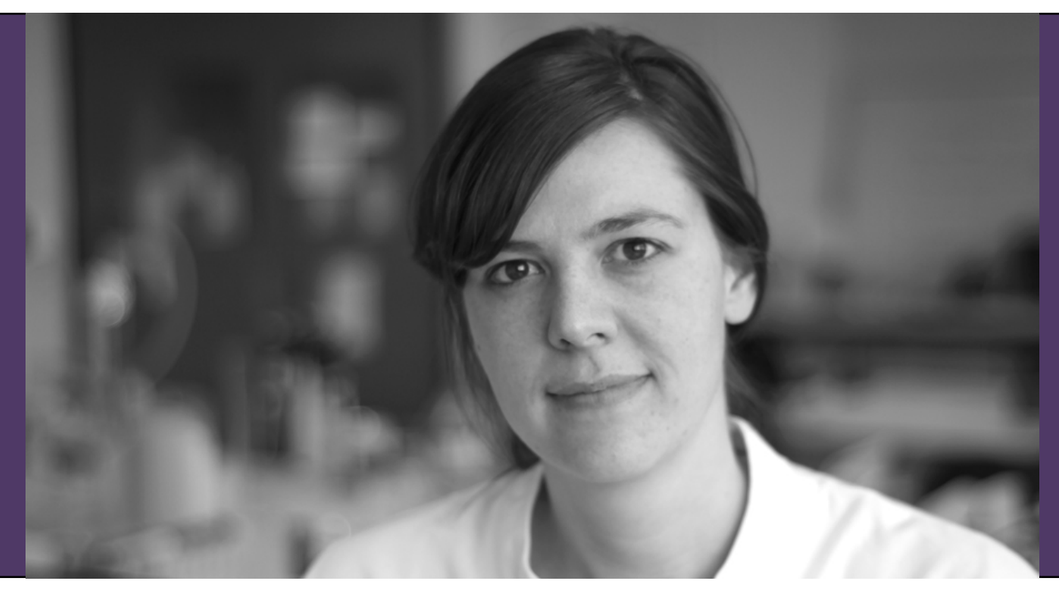

Introduction

Monoclonal antibodies (mAbs) dominate the biopharmaceuticals market. Of the 36 therapeutic mAbs commercially available or in review, 28 are IgG-1 and 8 are IgG-4 or IgG-2. These IgG mAbs target serious inflammatory conditions, cancers, autoimmune, cardiovascular and infectious diseases. Glycosylation can have significant effects on the clinical safety and efficacy of mAbs. As a result, therapeutic mAbs both biosimilar and innovator drugs, need to be fully characterized during the product lifecycle and in order to satisfy regulations.[1] Peptide mapping is a technique (Figure 1) which is routinely used to confirm the primary structure of proteins, is often used for the initial proof-of-structure characterization and is also heavily employed for lot-to-lot identity testing during bioprocess development.[2,3] The glycan analysis portion of glycoprotein characterisation is often completed alongside peptide mapping, however it generally involves an entirely separate series of specialised protocols.

At Ludger we have an active program for the development of mAb glycoprofiling technology to support modern biopharmaceutical design. We are interested in both the analysis of biologically relevant glycans and the corresponding techniques that allow us to do this analysis reliably and accurately. We work synergistically with biopharmaceutical companies to provide customised glycan analysis and also aid in the transfer of glycoprofiling methods to clients for in-house use. As a large proportion of our research programs are focused on the glycoprofiling of mAbs, we recognise when there is an opportunity for improvement or development. As mentioned above peptide mapping is routinely used in drug development and as part of the process both peptides and glycopeptides are generated. Thus, in an effort to streamline the characterisation process, we have developed the Velocity-Tag (V-Tag) System to add glycoprofiling to a peptide mapping workflow. Glycosylation analysis at the glycopeptide level is a favourable approach as site-specific glycan heterogeneity can be characterized and glycan compositions can be correlated to their attachment sites on the protein.

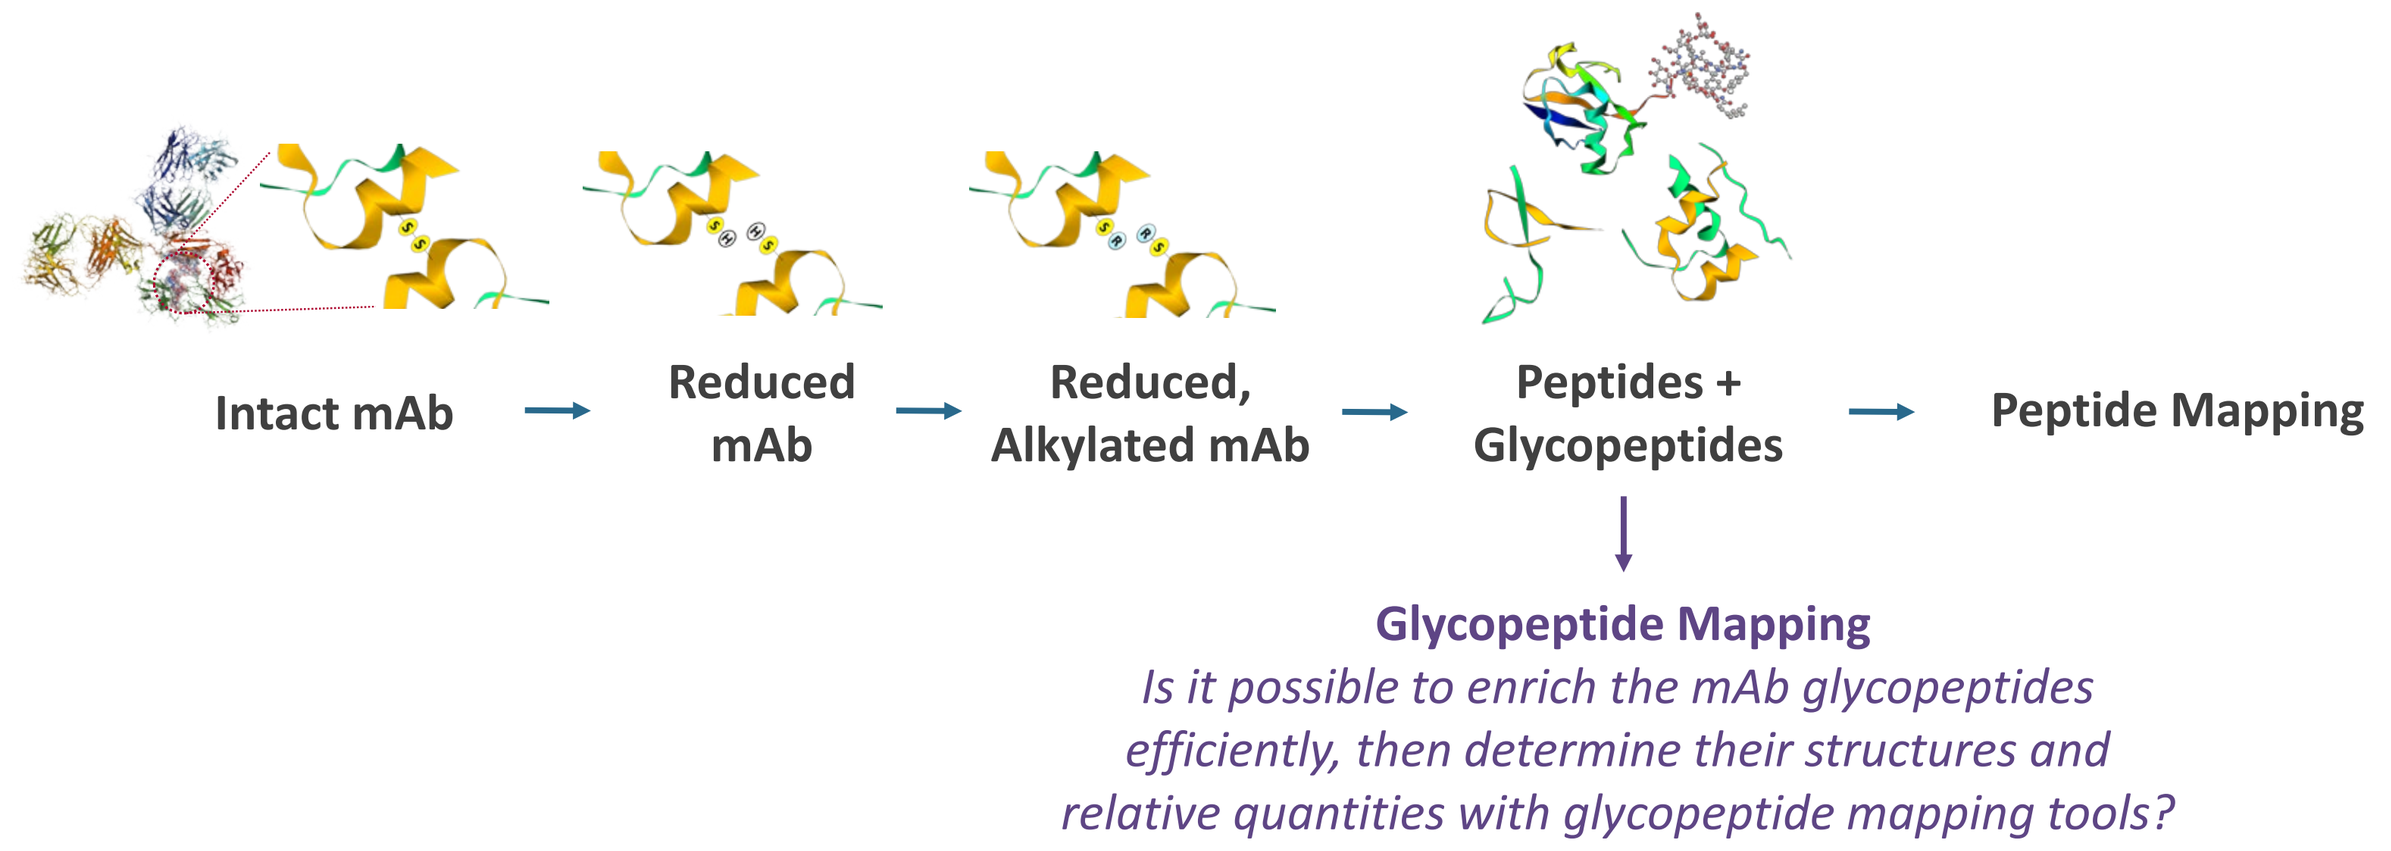
Intact mAb
Reduced mAb
Reduced, Alkylated mAb
Peptides + Glycopeptides
Peptide Mapping
Glycopeptide Mapping
Is it possible to enrich the mAb glycopeptides efficiently, then determine their structures and relative quantities with glycopeptide mapping tools?

Figure 1: Peptide mapping step-by-step procedure

LudgerTag V-Tag™ for Glycopeptide Mapping

The V-tag system is comprised of two steps which can be completed in 2 hours. The first step involves the labeling of peptides and glycopeptides in a protease digest using a novel fluorophore that has been synthetized from 2-amino-1-naphthalenesulfonic acid (Figure 2). The second step is the enrichment and clean-up of the labeled glycopeptides using a hydrophilic interaction liquid chromatography (HILIC) cartridge.

- **Integrates Easily with Peptide Mapping Workflow** - Adds onto your existing peptide mapping workflow, without requiring extra steps for glycan release
- **Minimal Sample Amount** - As little as 10 µg of glycoprotein (IgG) is required.
- **Enrichment of Glycopeptides** - Glycopeptides are enriched without degradation to preserve the glycosylation patterns and structures
- **Efficient and High Throughput** - labeling and glycopeptide enrichment is completed in 2 hours
- **Orthogonal Analysis** - V-Tag labeled glycopeptides can be analysed by MALDI-MS and (U)HPLC to give you reliable glycan identification and quantitation
- **Validated for GMP Labs** - Validated to ICH Q2(R1) standards and tested in GMP level glycoprofiling labs
- **Reliable mAb Glycoprofiling** - Provides data comparable to gold-standard glycoprofiling methods based on 2-AB or 2-AA
- **Automatable for High-Throughput Studies** - The procedure is compatible with 96-well plate based assays, enabling high-throughput studies using a liquid handling robot

Molecular Anatomy of V-Tag; An Amine Reactive Fluorescent label

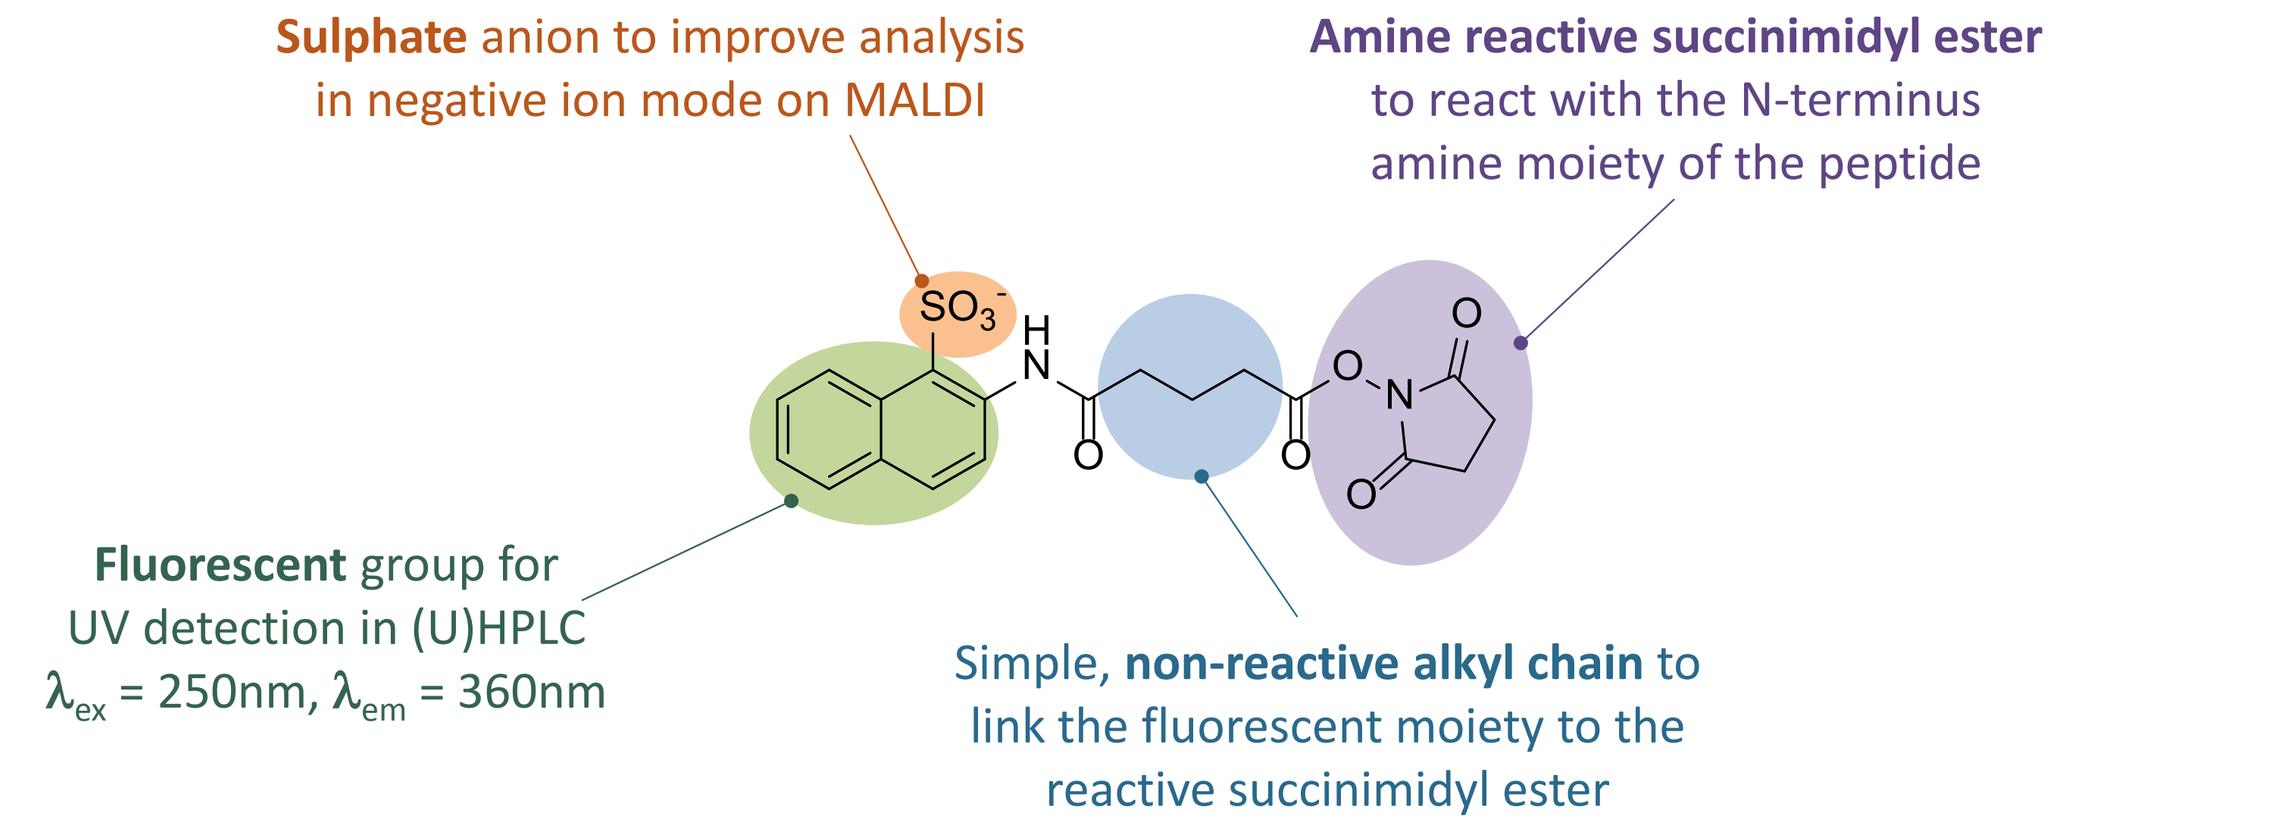
Sulphate anion to improve analysis in negative ion mode on MALDI
Amine reactive succinimidyl ester to react with the N-terminus amine moiety of the peptide
Fluorescent group for UV detection in (U)HPLC
λex = 250nm, λem = 360nm
Simple, non-reactive alkyl chain to link the fluorescent moiety to the reactive succinimidyl ester

Figure 2: V-Tag labeling Reagent

Methods

Detailed workflow for V-Tag labeling, enrichment and analysis of mAb glycopeptides

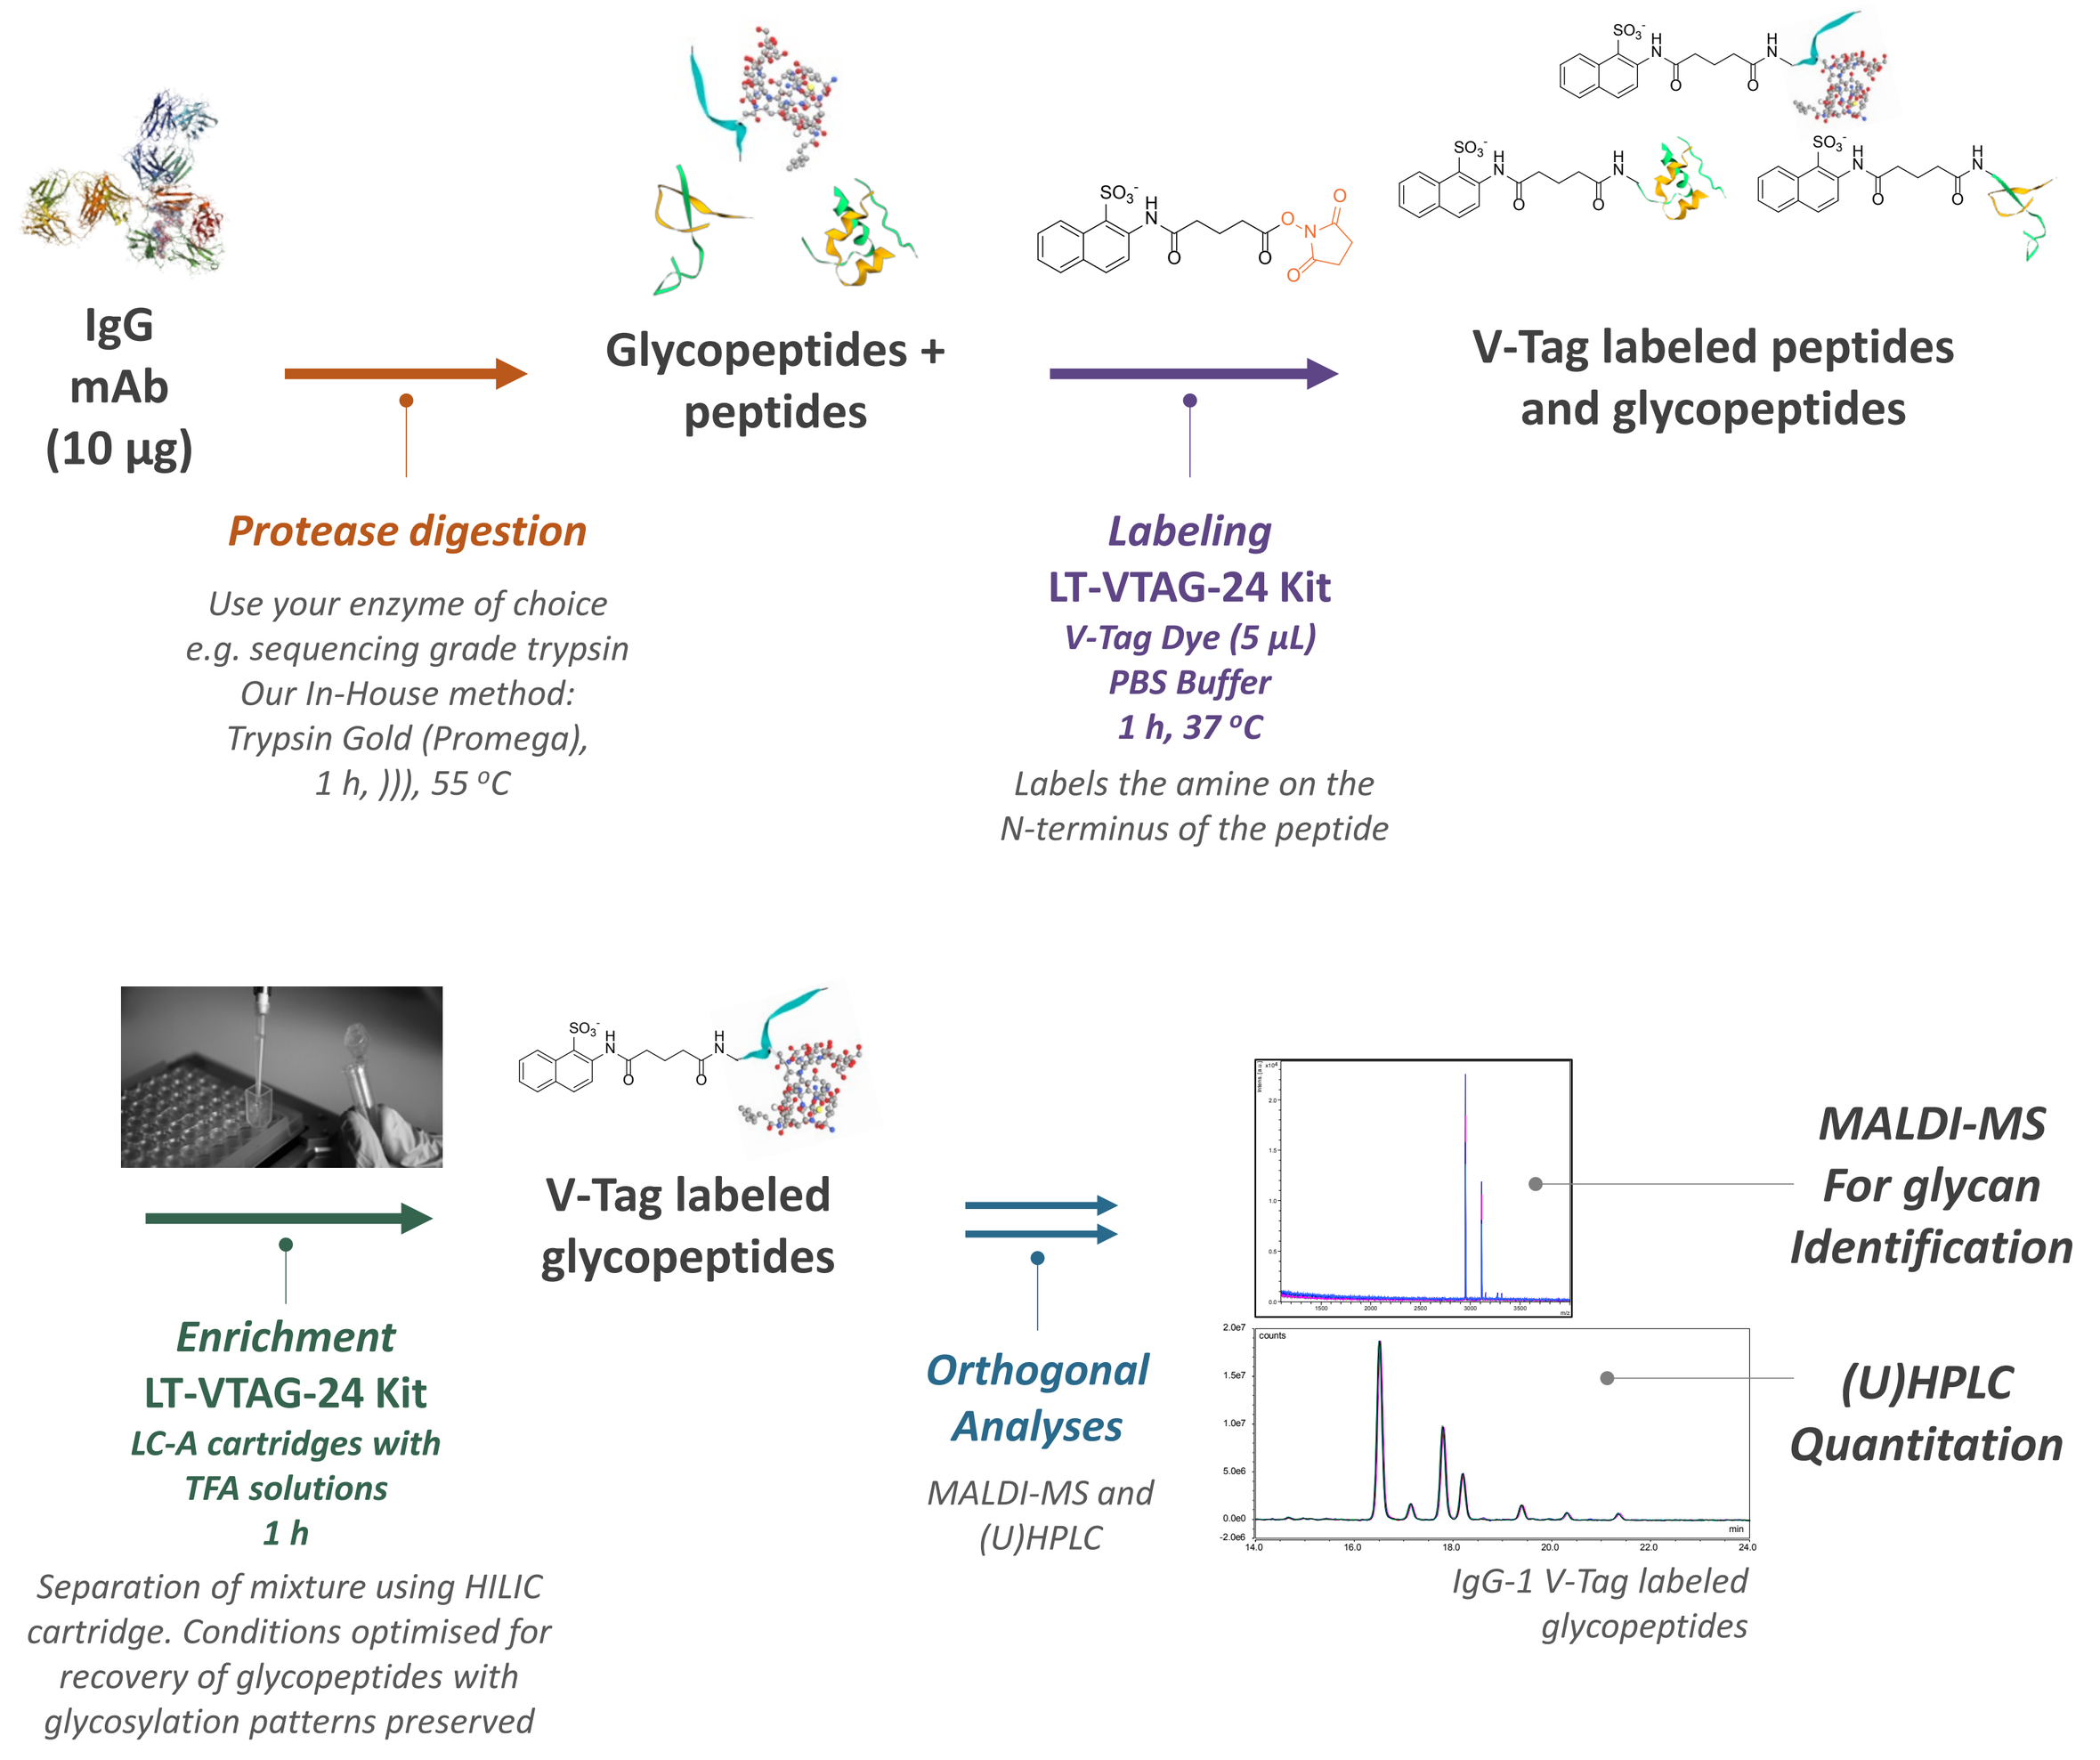
IgG mAb (10 µg)
Glycopeptides + peptides
V-Tag labeled peptides and glycopeptides
Protease digestion
Use your enzyme of choice
e.g. sequencing grade trypsin
Our In-House method:
Trypsin Gold (Promega),
1 h,))), 55 °C
Labeling
LT-VTAG-24 Kit
V-Tag Dye (5 µL)
PBS Buffer
1 h, 37 °C
Labels the amine on the N-terminus of the peptide
Enrichment
LT-VTAG-24 Kit
LC-A cartridges with TFA solutions
1 h
Separation of mixture using HILIC cartridge. Conditions optimised for recovery of glycopeptides with glycosylation patterns preserved
V-Tag labeled glycopeptides
Orthogonal Analyses
MALDI-MS and (U)HPLC
MALDI-MS For glycan Identification
(U)HPLC Quantitation
IgG-1 V-Tag labeled glycopeptides

Analysis of V-Tag labeled glycopeptides

For detailed glycan characterization we use a combination of two orthogonal analyses: MALDI-MS and HILIC (Hydrophilic Interaction Liquid Chromatography) (U)HPLC. These provide two essential drug characterization parameters which are glycan identification (from the MALDI-MS) and the relative molar quantitation (from the (U)HPLC).

(U)HPLC

Typical setup for analysis of V-Tag labeled glycopeptides by (U)HPLC

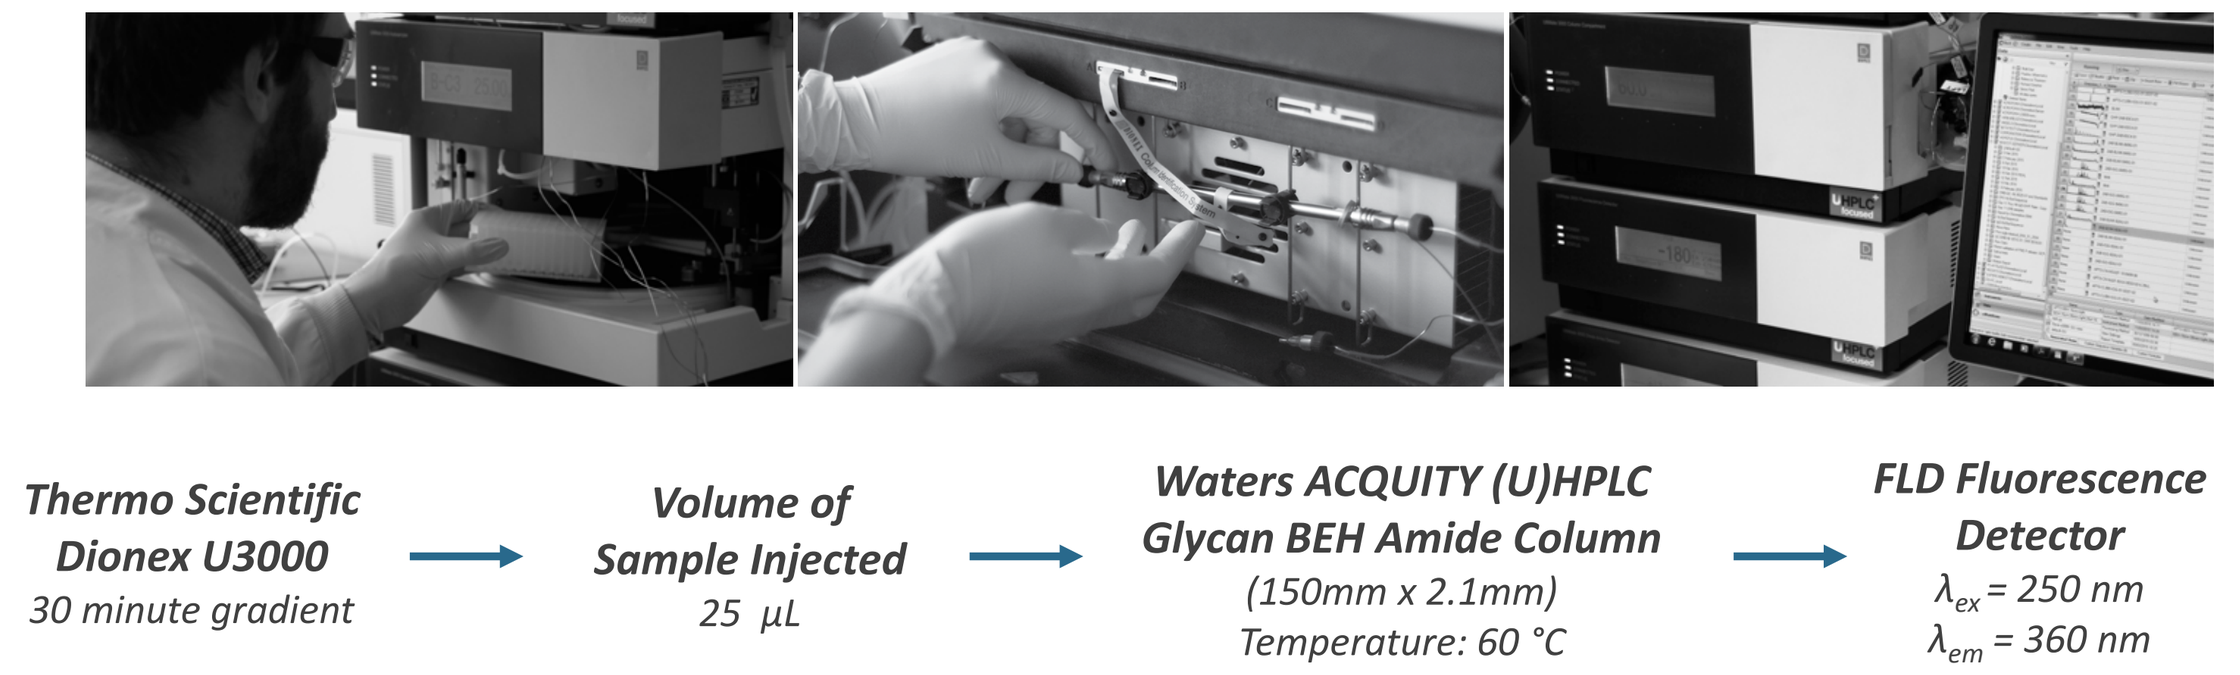
Thermo Scientific Dionex U3000
30 minute gradient
Volume of Sample Injected
25 µL
Waters ACQUITY (U)HPLC Glycan BEH Amide Column
(150mm x 2.1mm)
Temperature: 60 °C
FLD Fluorescence Detector
λex = 250 nm
λem = 360 nm

MALDI-MS

Typical setup for analysis of V-Tag glycopeptides by MALDI-MS

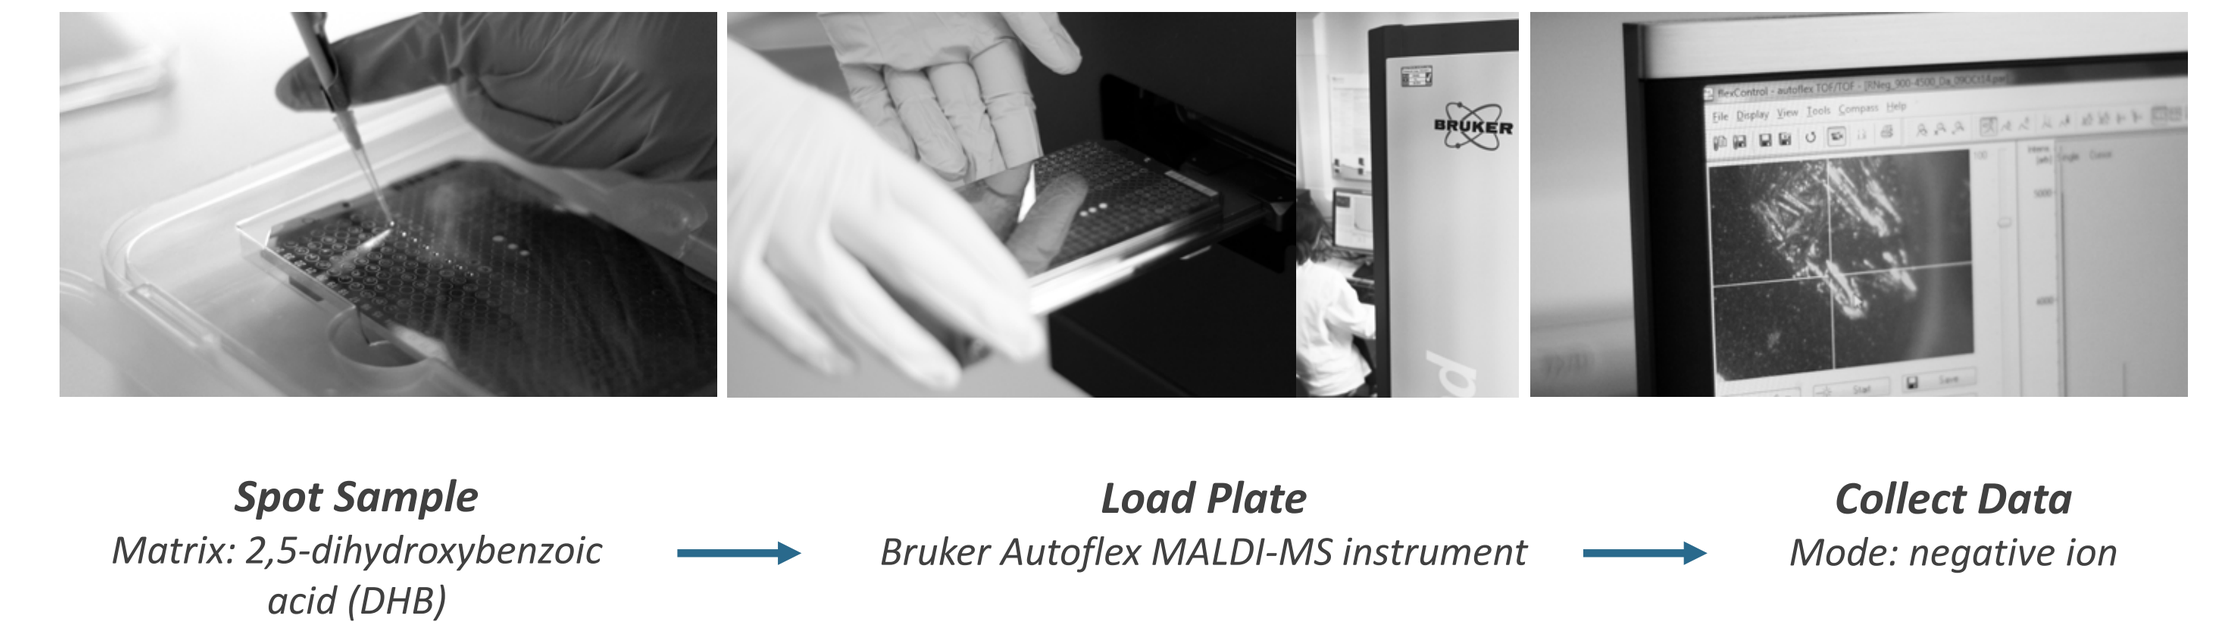
Spot Sample
Matrix: 2,5-dihydroxybenzoic acid (DHB)
Load Plate
Bruker Autoflex MALDI-MS instrument
Collect Data
Mode: negative ion

Results and Discussion

Analysis of Tryptic IgG-1 mAb Glycopeptides Using V-Tag labeling and Enrichment

i) MALDI-MS for Identification

MALDI-MS analysis of the IgG tryptic digest was completed both before and after V-Tag labeling and enrichment. The MS trace for the IgG tryptic digest shows ion signals for both peptides and glycopeptides, with the glycopeptides having a low signal intensity (Figure 3A, glycopeptide region in dotted box). In comparison, the MS trace acquired after V-tag labeling and enrichment shows both a good enrichment of glycopeptides and an increase in ion signal intensity (Figure 3B). MALDI-MS ion signals were assigned by calculating the corresponding glycan composition for each m/z based on known IgG N-glycopeptide structures (Figure 4).

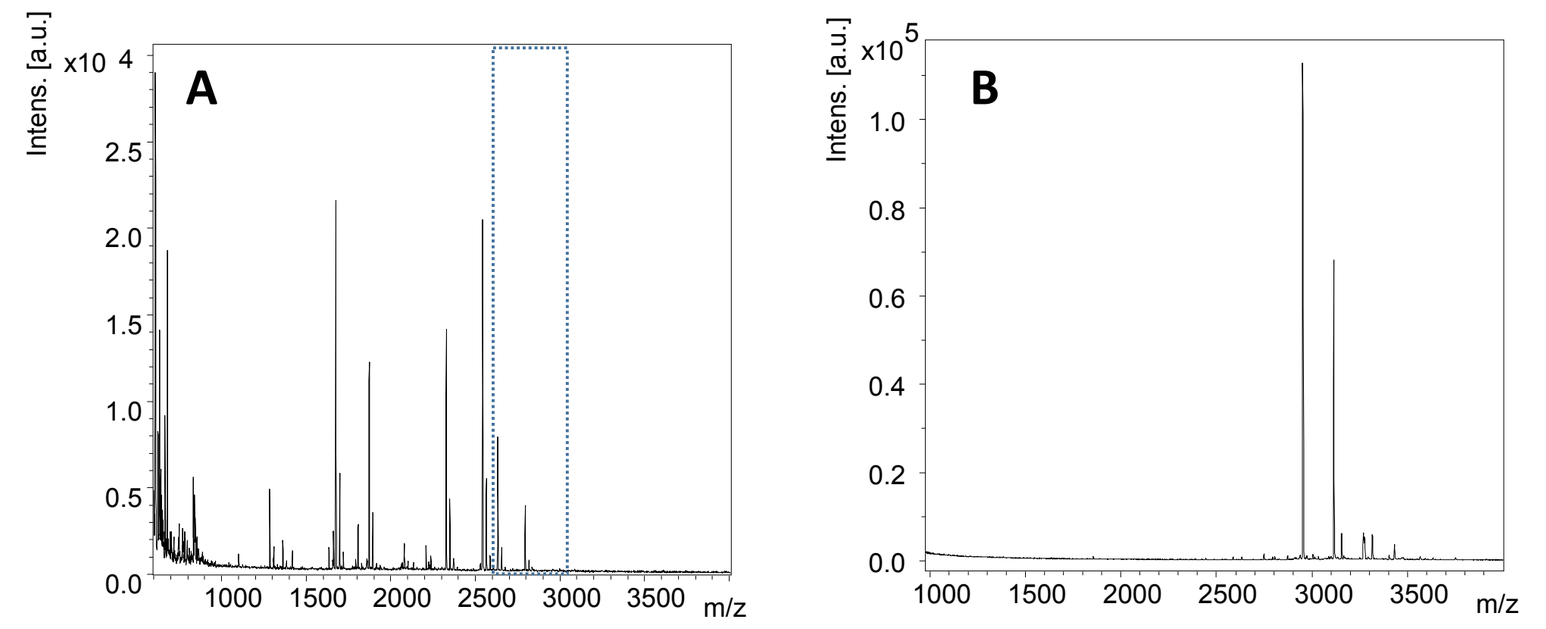
A
B
Intens. [a.u.]
m/z

Figure 3: A) MALDI-MS trace of IgG mAb tryptic digest (negative ion mode)
B) MALDI-MS trace of IgG mAb glycopeptides after V-Tag labeling and enrichment (negative ion mode)

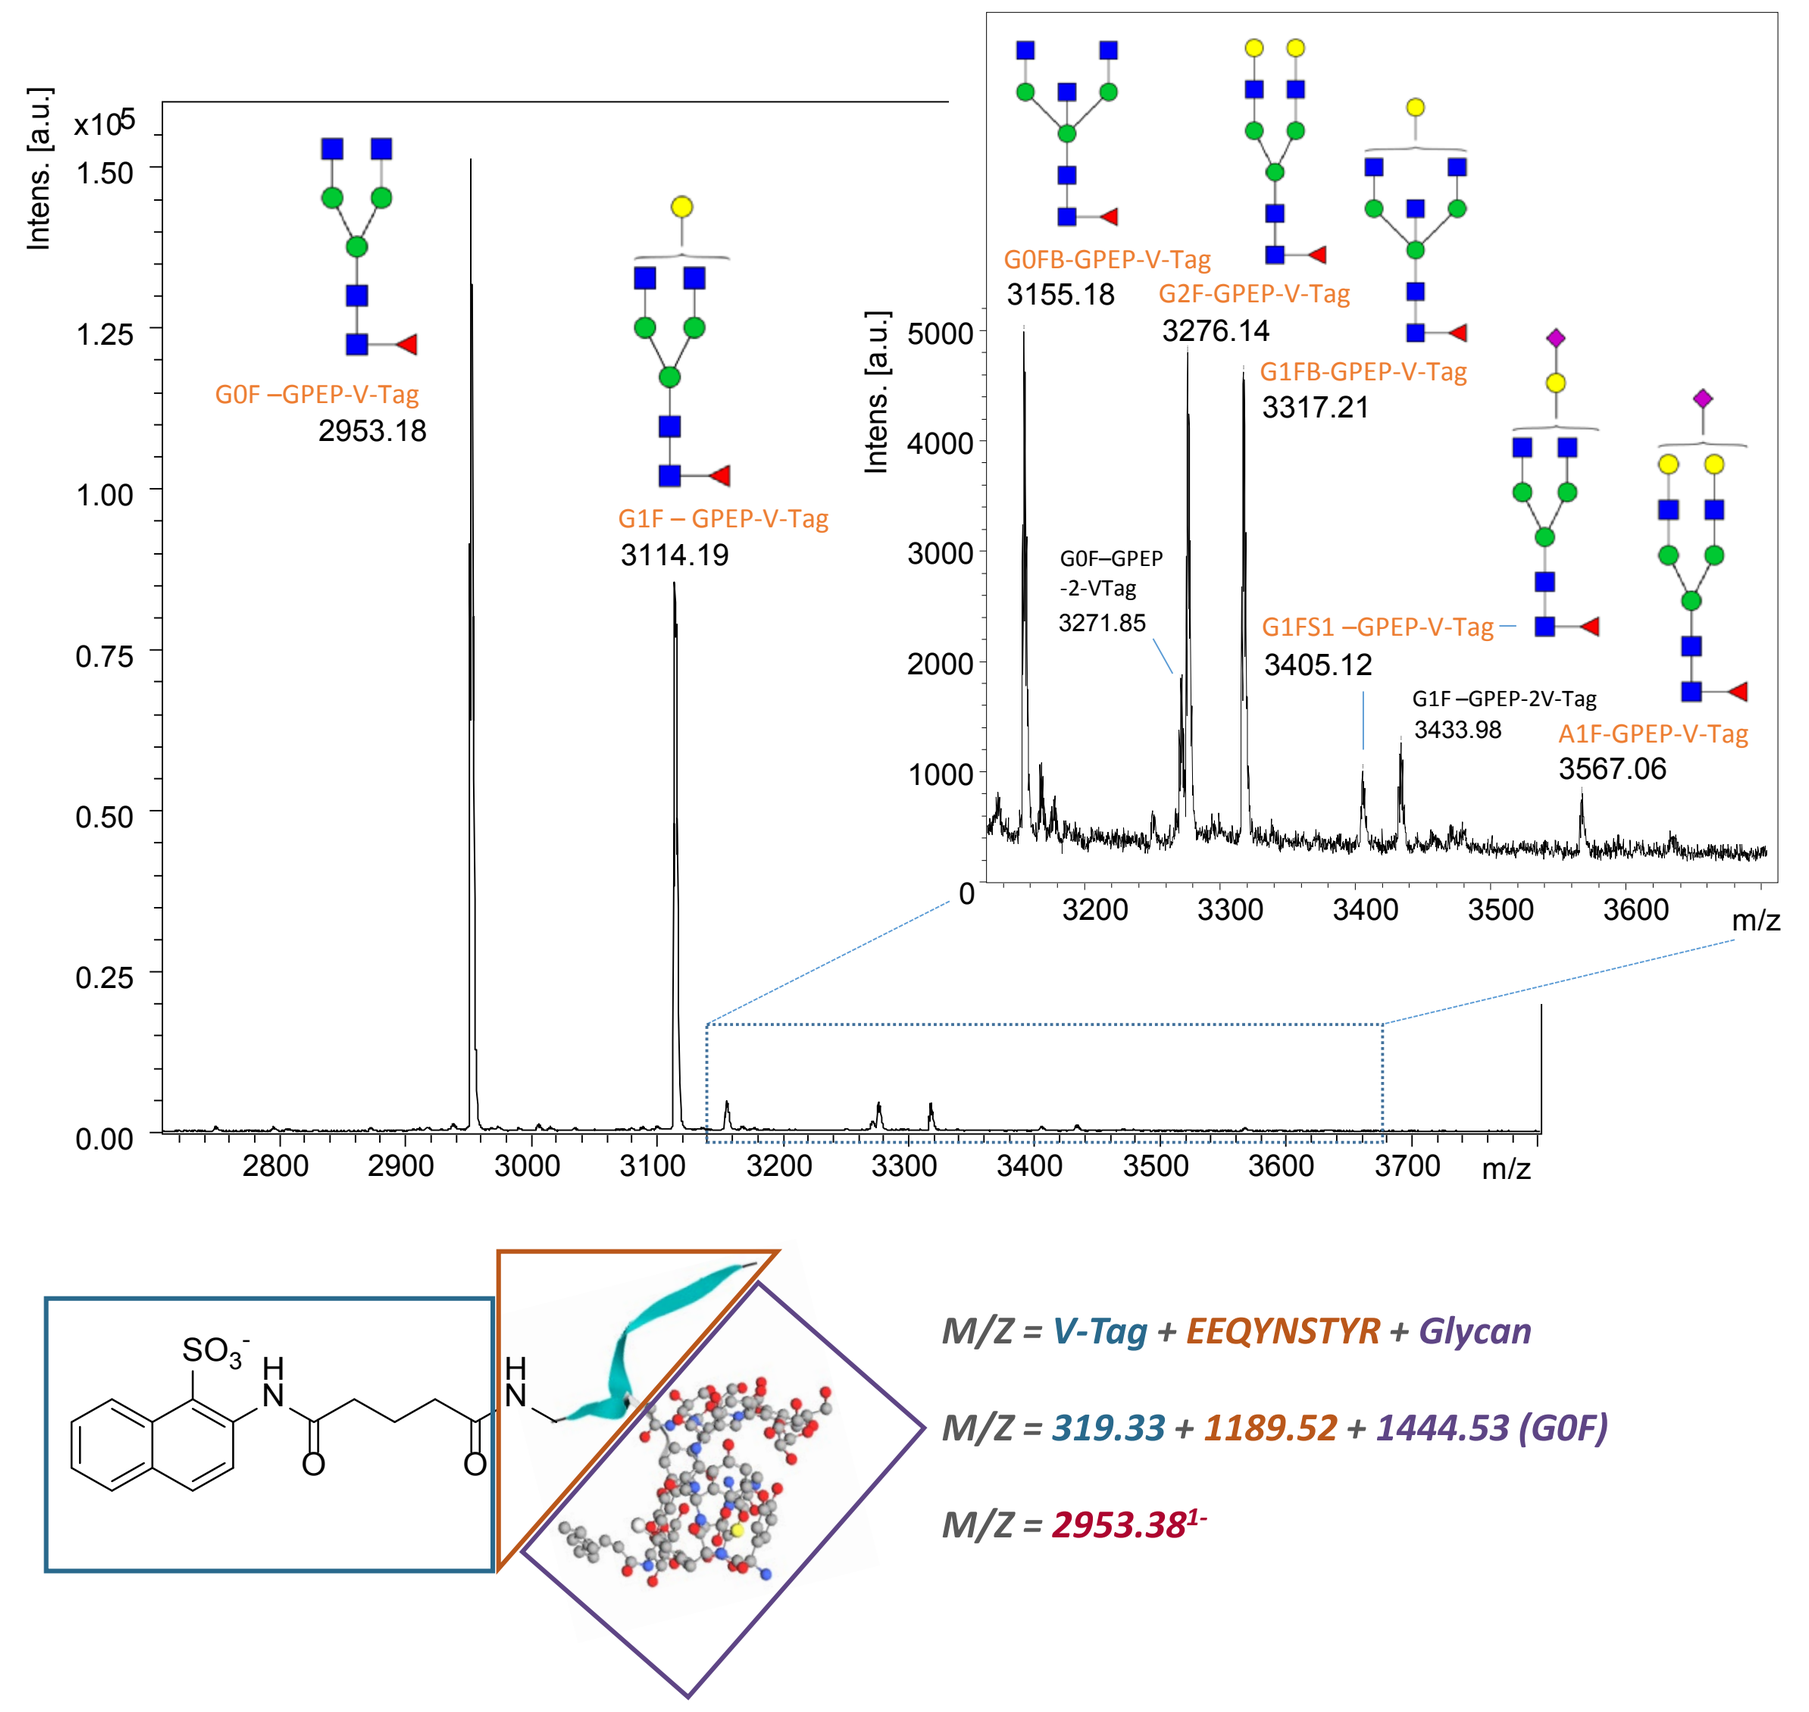
G0F –GPEP-V-Tag
2953.18
G1F – GPEP-V-Tag
3114.19
G0FB-GPEP-V-Tag
3155.18
G2F-GPEP-V-Tag
3276.14
G1FB-GPEP-V-Tag
3317.21
G0F–GPEP -2-VTag
3271.85
G1FS1 –GPEP-V-Tag
3405.12
G1F –GPEP-2V-Tag
3433.98
A1F-GPEP-V-Tag
3567.06
M/Z = V-Tag + EEQYNSTYR + Glycan
M/Z = 319.33 + 1189.52 + 1444.53 (G0F)
M/Z = 2953.38¹⁻

Figure 4: MALDI-MS trace for V-tag labeled IgG glycopeptides and how to calculate M/Z

ii) (U)HPLC for quantitation

The relative quantitation for the various V-tag labeled N-glycopeptide IgG isoforms was calculated from the (U)HPLC chromatograms. In addition to reproducible qualitative results (Figure 5, overlay of chromatograms (n=9)), the V-Tag labeling and enrichment of IgG glycopeptides gave quantitative results with good reproducibility having CVs with less than 4% (Table 1). The V-Tag system has been validated to ICH Q2(R1) level.

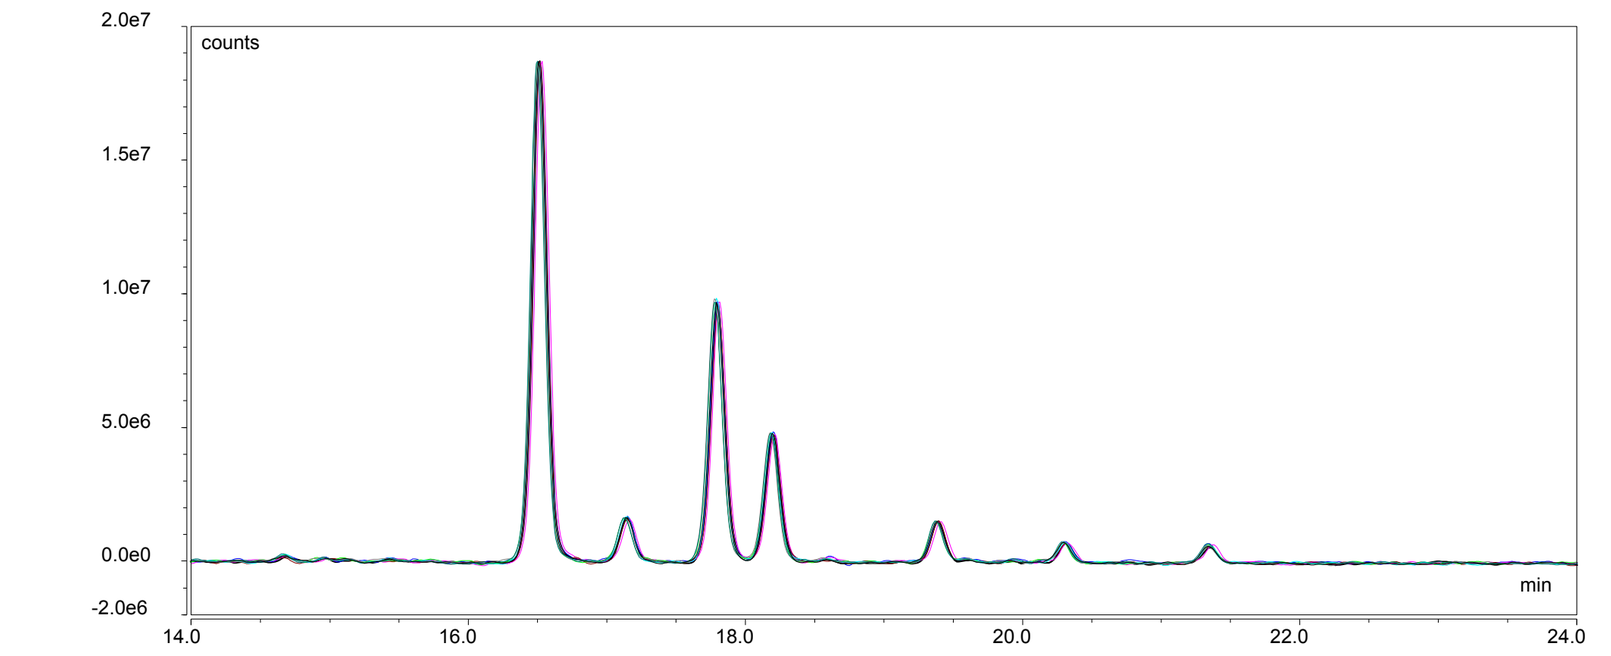

Figure 5: Overlaid (U)HPLC chromatograms (n=9) of V-tag labeled glycopeptides from IgG

Peak Number		1	2	3	4	5	6	7
Glycopeptide		G0F	G0FB	G1F	G1F + G1FB	G2F	G1FS1	A1F
Relative % Area	Av.	50.22	4.22	25.83	12.58	3.80	1.80	1.73
	Std. Dev.	0.35	0.14	0.18	0.26	0.10	0.02	0.04
	CV	0.70	3.20	0.69	2.05	2.54	0.92	2.40

Table 1: Relative abundance and reproducibility for V-tag labeled glycopeptides from IgG (n=9)

iii) Comparison of 2-AB and V-Tag Glycoprofiling by (U)HPLC

Figure 6 shows a comparison of the methods for the gold standard method of in-solution PNGase F digest followed by 2-AB labelling of N-gycans and the V-Tag labeling and enrichment of N-glycopeptides. A representative chromatogram from each method shows that V-Tag labeling of N-glycopeptides gives essentially the same profile as the standard 2-AB labeling of N-glycans. The relative abundance of the V-Tag labeled glycopeptides obtained from trypsin digestion compares with the relative abundance obtained with the orthogonal, 2-AB labeling N-glycans (3 replicates each, Figure 7). Thus, incorporating glycoprofiling into the peptide mapping workflow with V-Tag offers a reliable and efficient alternative to N-glycan release and labeling.

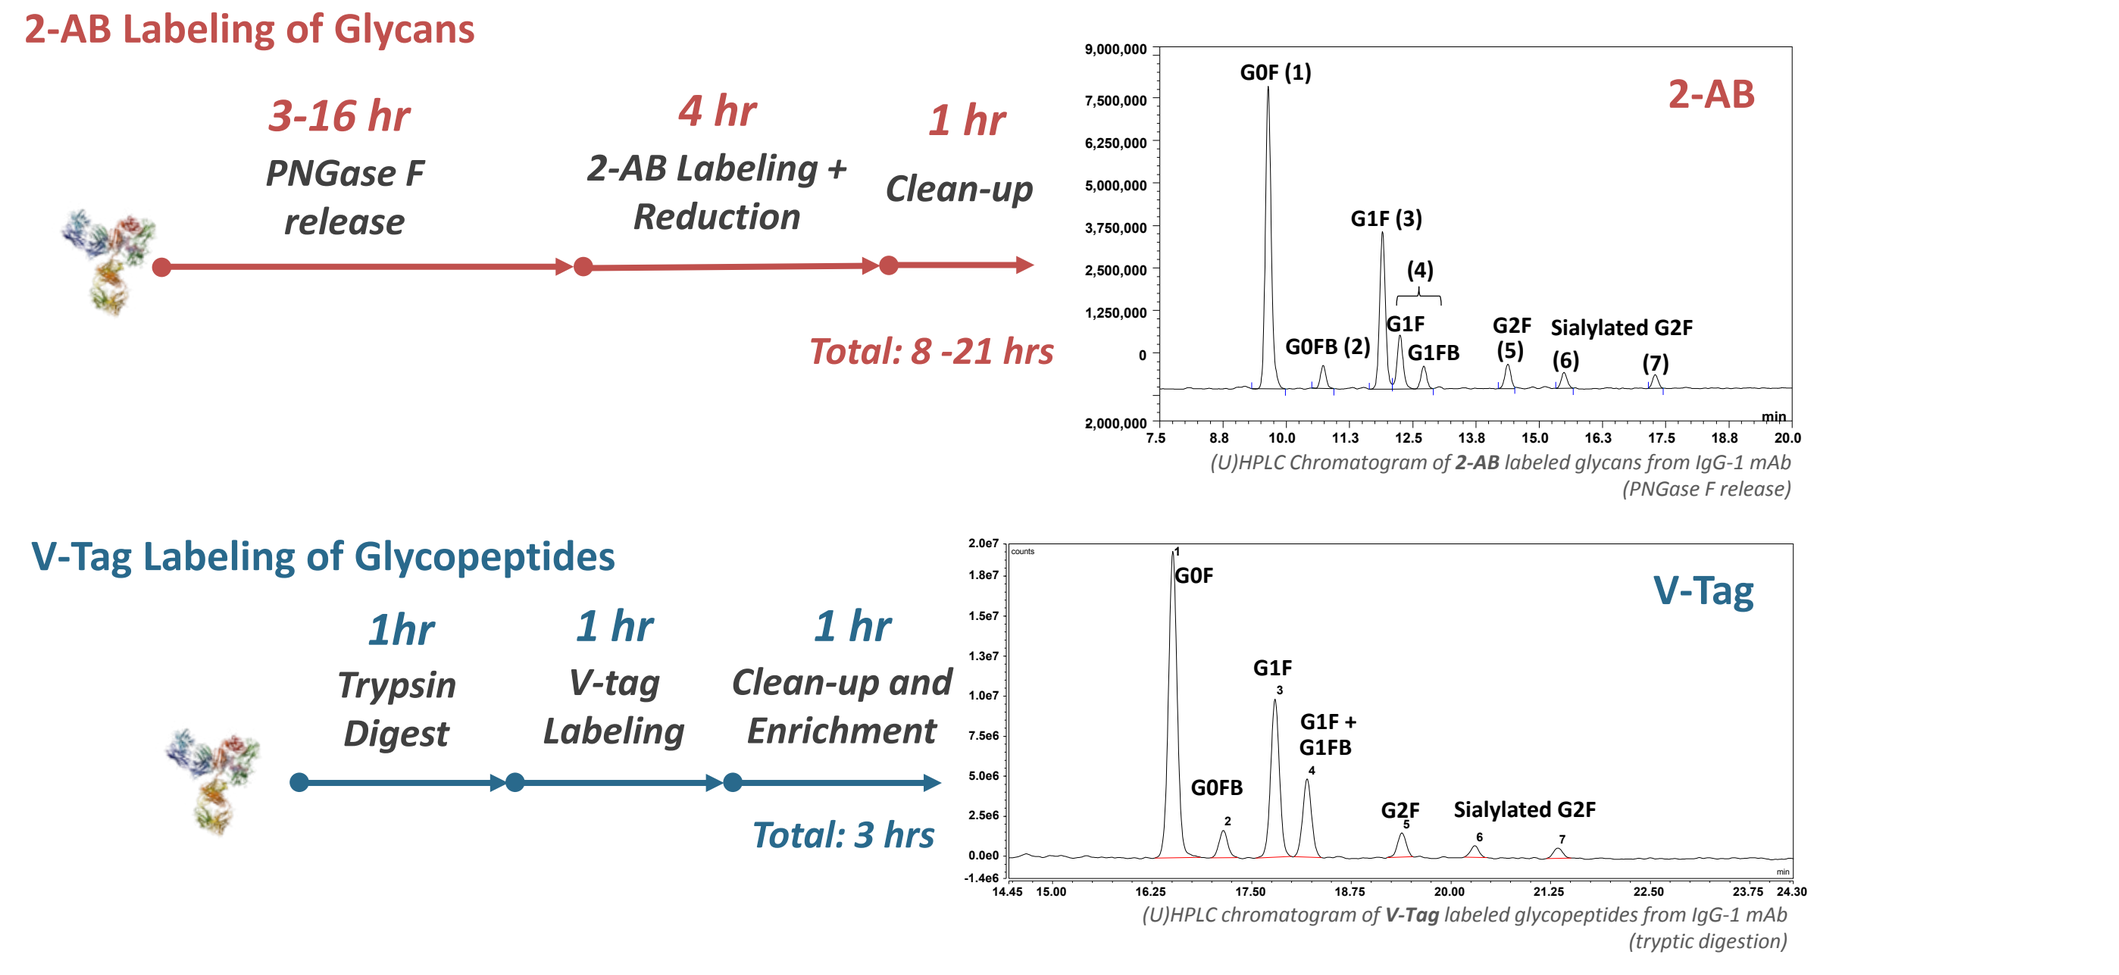
2-AB Labeling of Glycans
3-16 hr
PNGase F release
4 hr
2-AB Labeling + Reduction
1 hr
Clean-up
Total: 8 -21 hrs
2-AB
G0F (1)
G1F (3)
G0FB (2)
G1F
G1FB
G2F (5)
Sialylated G2F (6) (7)
(U)HPLC Chromatogram of 2-AB labeled glycans from IgG-1 mAb (PNGase F release)
V-Tag Labeling of Glycopeptides
1hr
Trypsin Digest
1 hr
V-tag Labeling
1 hr
Clean-up and Enrichment
Total: 3 hrs
V-Tag
G0F
G0FB
G1F
G1F + G1FB
G2F
Sialylated G2F
(U)HPLC chromatogram of V-Tag labeled glycopeptides from IgG-1 mAb (tryptic digestion)

Figure 6: Workflow and UHPLC chromatograms for 2-AB and V-Tag Glycoprofiling

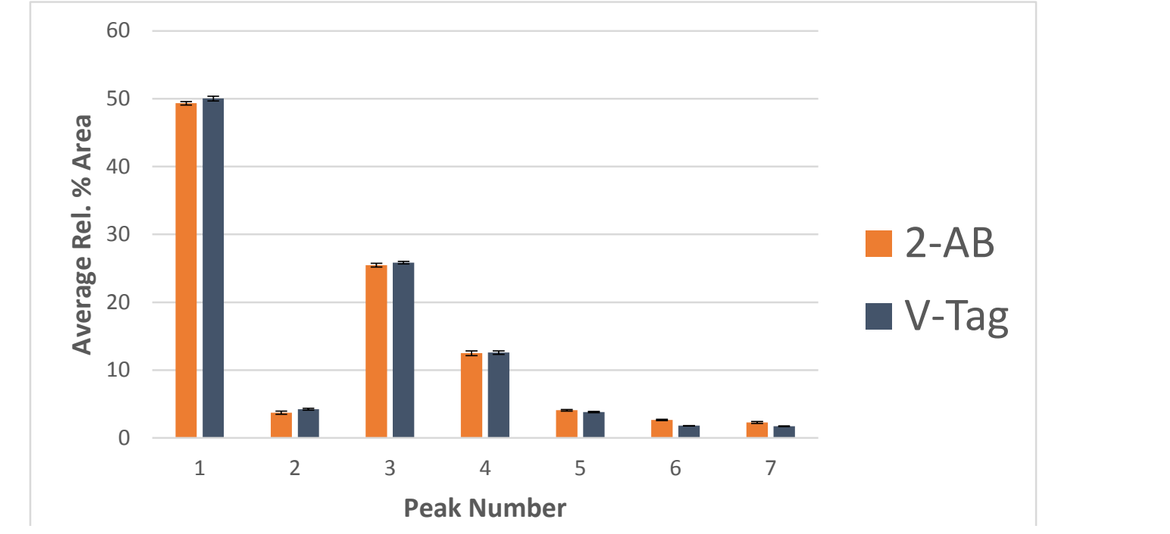
Average Rel. % Area
Peak Number
2-AB
V-Tag

Figure 7: The relative abundance of the V-Tag labeled trypitic N-glycopeptides compares with the relative abundance of PNGaseF released, 2-AB labeled N-Glycans

iv) Study of IgG mAb Glycosylation in Fermentation Supernatants; An Example of choosing cell culture sparging conditions to optimise Fc galactosylation

Chinese hamster ovary (CHO) cell line GS-CY01 expressing an IgG mAb was grown in bio-reactors using different aeration conditions.[4] The V-tag system was used to investigate the differences in Fc galactosylation (G0F, G1F and G2F). MALDI-MS and (U)HPLC data was collected for all of the sparging conditions (Figure 8).

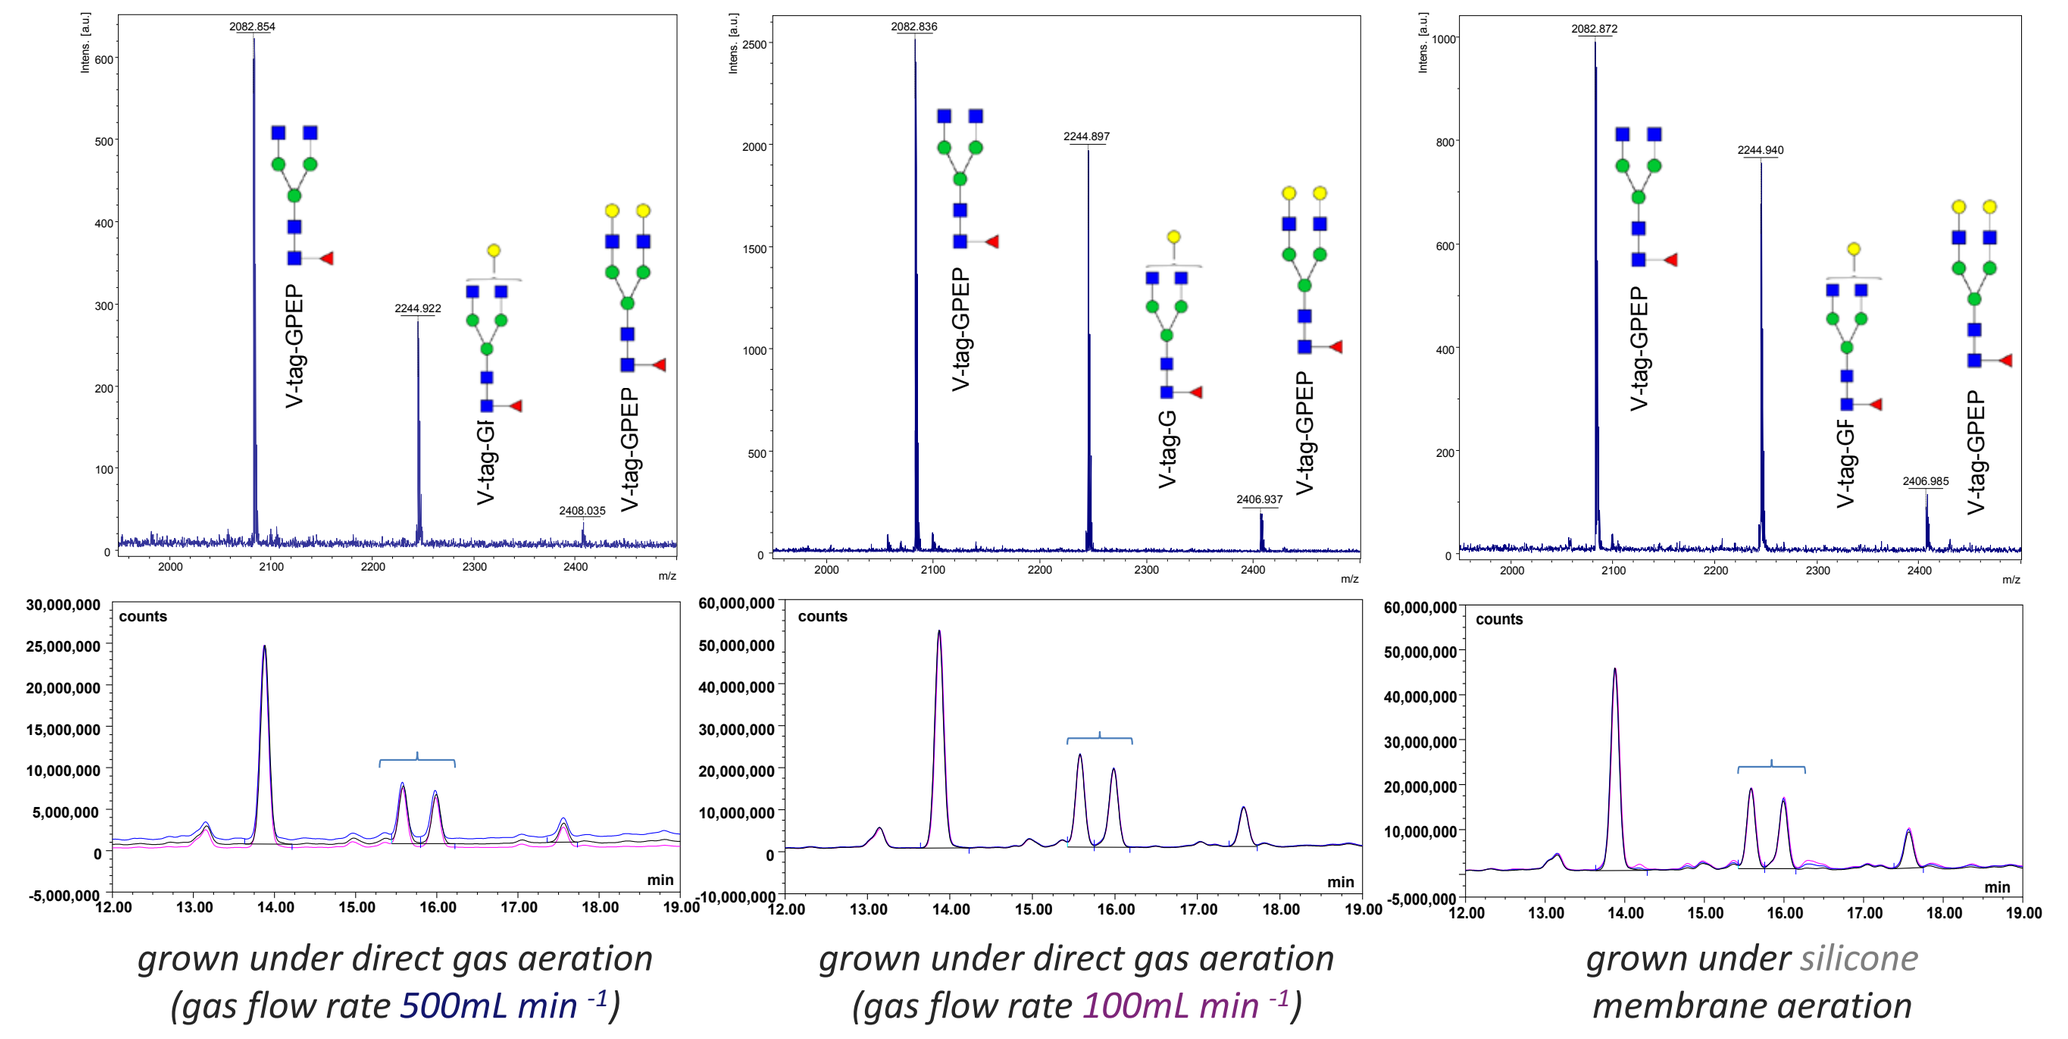
grown under direct gas aeration (gas flow rate 500mL min⁻¹)
grown under direct gas aeration (gas flow rate 100mL min⁻¹)
grown under silicone membrane aeration

Figure 8: MALDI-MS traces and UHPLC chromatograms for culture sparging conditions

The Fc galactosylation patterns (i.e. the ratios of the G0F, G1F and G2F glycans) changed according to the type of bioreactor aeration (Figure 9). The cells grown under silicon membrane aeration showed the highest degree of Fc galactosylation (higher abundance of G2F). Fc galactosylation may impact the mAb complement dependent cytotoxicity (CDC) as increasing the levels of terminal galactose are known to positively correlate with CDC activity.[5] This study illustrates that subtle variations in glycoform patterns are reliably detected with the V-Tag System making it a viable method for QbD (Quality by Design) studies in drug development and for optimisation of mAb glycosylation.

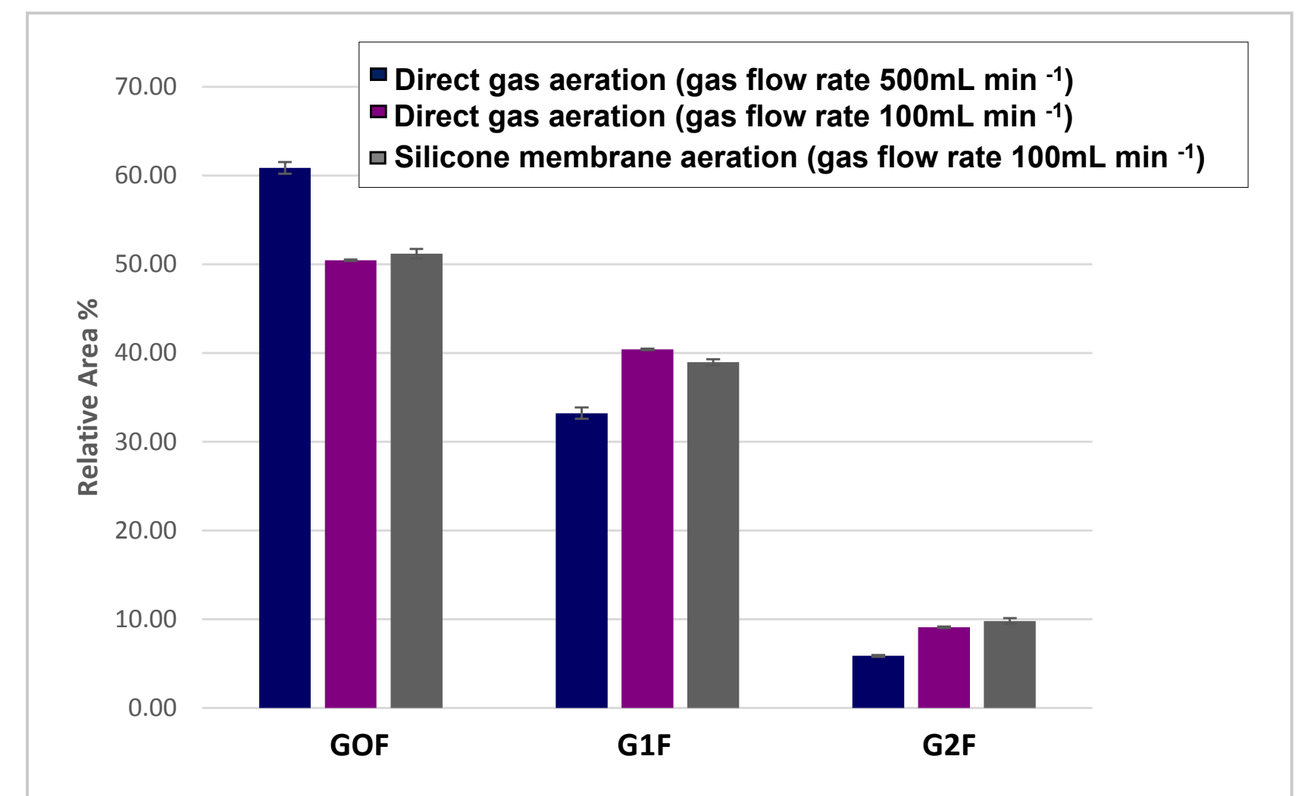
Direct gas aeration (gas flow rate 500mL min⁻¹)
Direct gas aeration (gas flow rate 100mL min⁻¹)
Silicone membrane aeration (gas flow rate 100mL min⁻¹)
Relative Area %
GOF
G1F
G2F

Figure 9: The ratios of relative abuncance for G0F, G1F and G2F glycans change according to the type of bioreactor aeration

v) Automated High Throughput Glycomics Studies using V-Tag

We have adapted the V-Tag workflow to a 96-well plate system to allow for its use with a liquid handling robot. The workflow (Figure 10) can be completed in a day which makes this technology is a good candidate for high throughput analysis of mAbs.

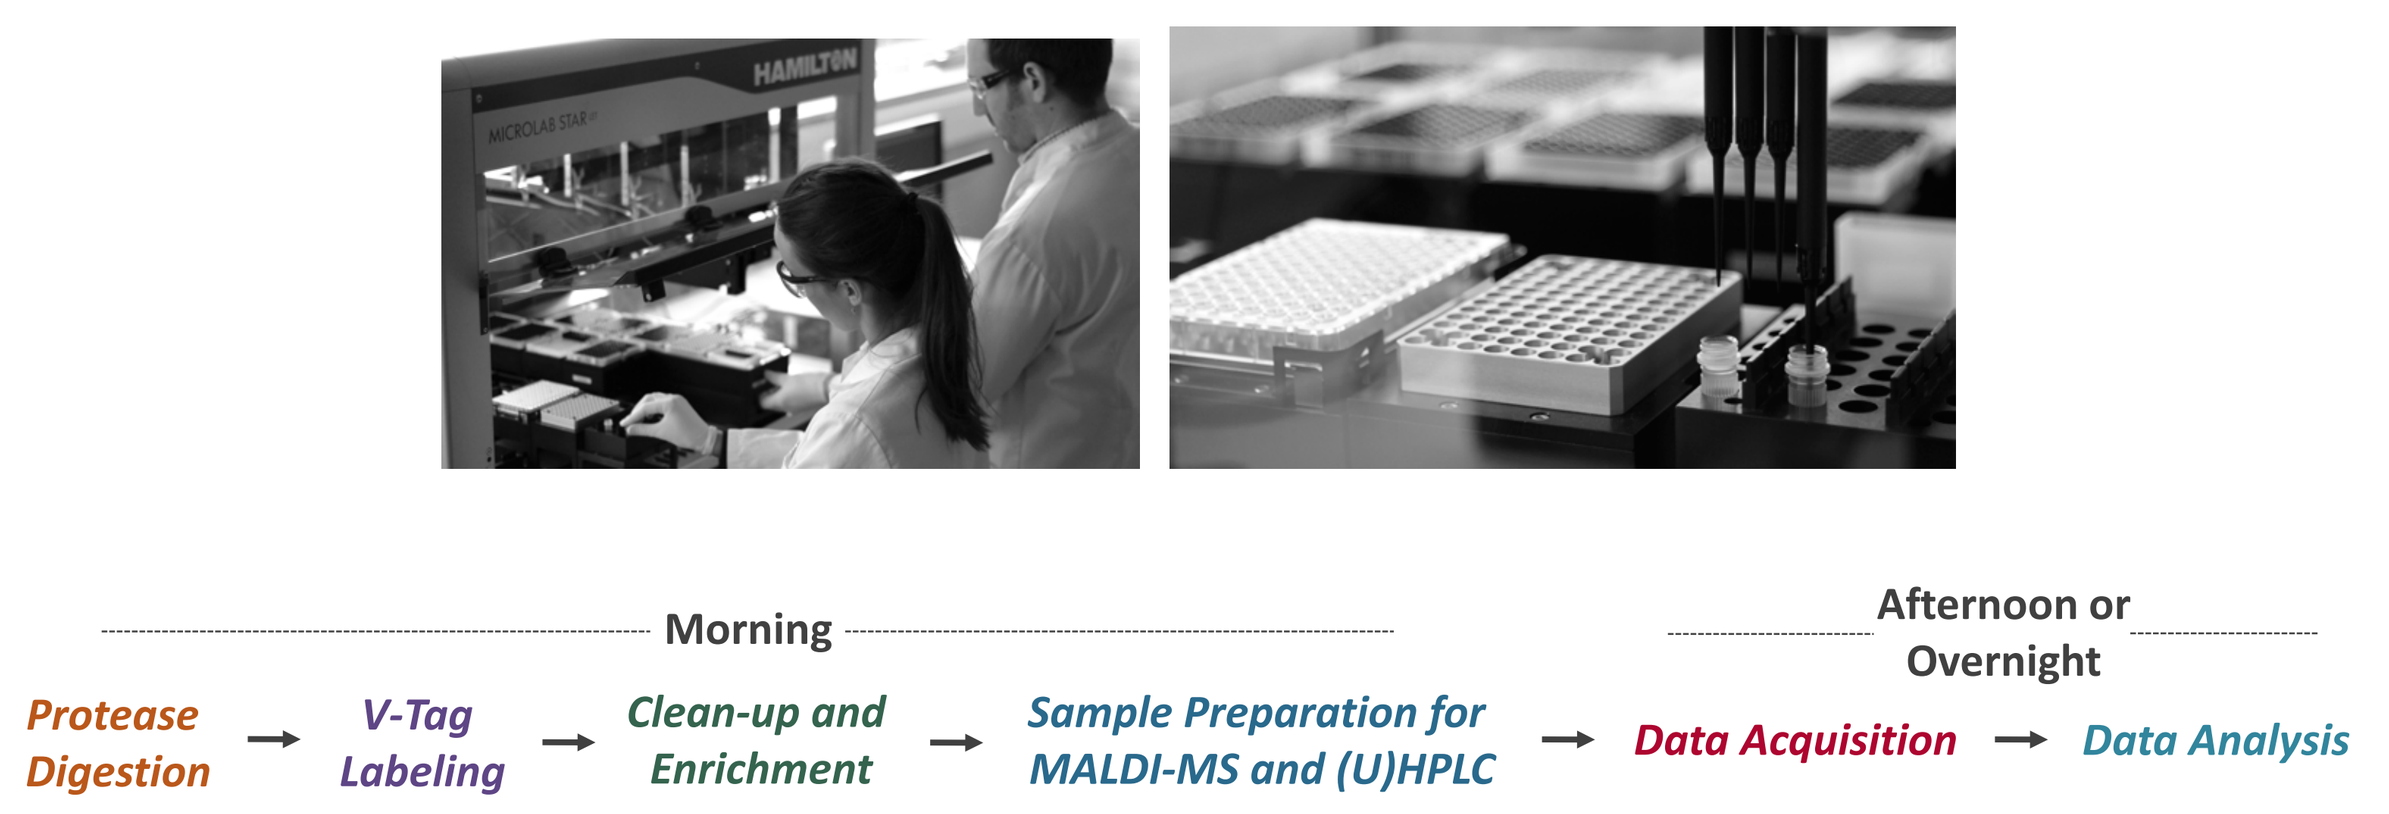
Morning
Afternoon or Overnight
Protease Digestion
V-Tag Labeling
Clean-up and Enrichment
Sample Preparation for MALDI-MS and (U)HPLC
Data Acquisition
Data Analysis

Figure 10: Automated V-Tag workflow can be completed in 1 day

Conclusions

- The V-Tag labeling and enrichment system for IgG mAb glycopeptides affords reliable glycan identification and quantitation data using orthogonal methods (MALDI-MS and (U)HPLC) and integrates easily into the peptide mapping workflow
- The enrichment of glycopeptides allows for an increased signal intensity on MALDI-MS.
- The V-Tag labeling of N-glycopeptides yields data that is comparable to 2-AB labelling of the corresponding N-glycans.
- The V-Tag system has been used successfully to study the variation in glycosylation profiles for IgG mAbs grown in different bio-reactors with different aeration conditions.
- The V-Tag workflow has been adapted to a 96-well plate system to allow for its use with a liquid handling robot, making this technology a good candidate for high throughput analysis of IgG mAbs.

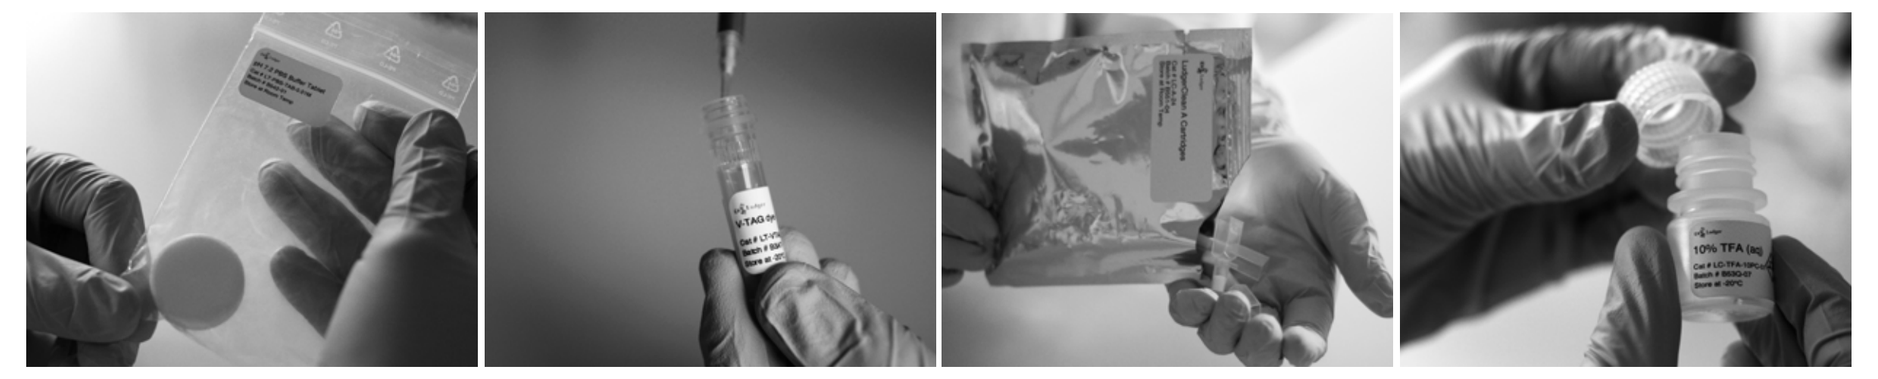

If you would like more information about the V-Tag Glycopeptide Labeling and Enrichment System or would like to submit a sample for analysis as part of our glycoprofiling service (we perform the analysis for you in our GMP level glycoprofiling labs and send you a data report): Contact Jenifer

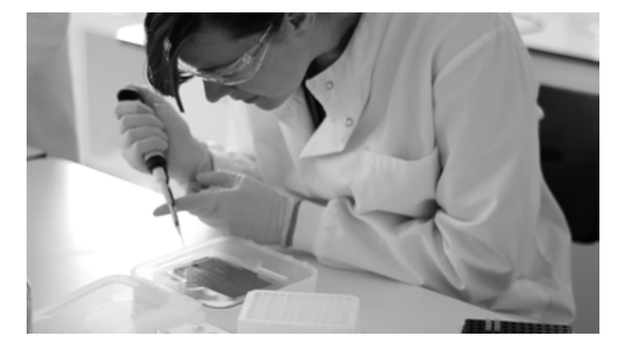

Jenifer Hendel
Scientist
jenifer.hendel@ludger.com

If you would like to try V-Tag in your lab: Contact Karen for a quotation (Catalogue # LT-VTAG-24)

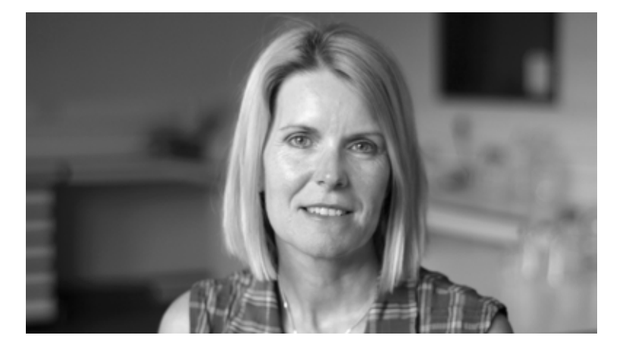

Karen Oakes
Sales Manager
sales@ludger.com

Acknowledgements

D. G. Bracewell at University College London for IgG from supernatents
Matthew Doherty at Ludger Ltd. for poster design

References

1. L. Liming, *J. Pharm. Sci.*, 2015, 1866–1884.
2. J. Bongers *et al.* *J. Pharm. Biomed. Anal.* **2000**, 21, 1099-1128.
3. 'peptide mapping' search word on http://www.pharmacopeia.cn
4. A. S. Tait, R. D. R. Tarrant, M. L. Velez-Suberbie, D. I. R. Spencer, D. G. Bracewell, *Biotechnol. Prog.* **2013**, 29, 688-696.
5. T. S. Raju, R. Jordan *mAbs*, **2012**, *4*, 385-391.